DISCOVERY OF NOVEL SMALL MOLECULES FOR THE TREATMENT OF FTD-*GRN*

Christopher J. Holler, Angela Chen, Natasha Khatri, James C. Lanter, Ricardo L. Sanz, Jerusha K. Brendel, Mary Wozniak, Duane A. Burnett, Gerhard Koenig, Jean-François Blain

Arkuda Therapeutics, Watertown, MA, 02472

Background and Approach:

Heterozygous or homozygous mutations in the *GRN* gene, encoding progranulin (PGRN), cause frontotemporal dementia (FTD) or neuronal ceroid lipofuscinosis (NCL), respectively. FTD-*GRN* and NCL-*GRN* are characterized by lysosome dysfunction and neurodegeneration, indicating PGRN is important for lysosome homeostasis in the brain. PGRN can be secreted or trafficked to the lysosome where it is processed into smaller proteins called granulins which are believed to be the functional units of PGRN. Since PGRN and granulins are deficient in FTD-*GRN*, strategies aimed at increasing their secretion (PGRN) or lysosomal content (granulins) in the brain are a rational and attractive therapeutic approach to a disease with no currently approved treatments.

Role of Progranulin in the Brain

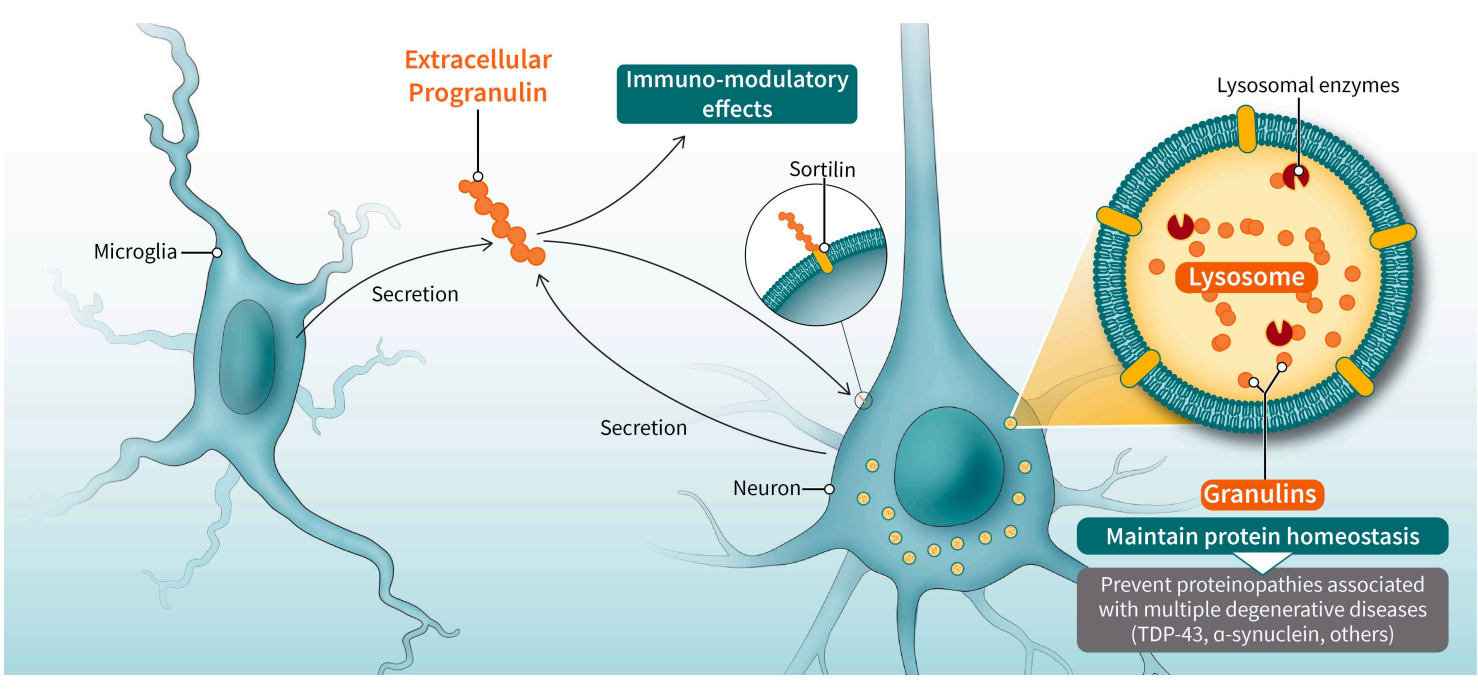
Extracellular Progranulin
Immuno-modulatory effects
Lysosomal enzymes
Sortilin
Microglia
Secretion
Lysosome
Secretion
Neuron
Granulins
Maintain protein homeostasis
Prevent proteinopathies associated with multiple degenerative diseases (TDP-43, α-synuclein, others)

Here, we describe a small molecule screen in BV-2 cells with the aim of identifying novel molecules to increase both progranulin secretion and lysosomal granulins. Through structure activity relationship optimization, we improved the potency of compounds from >2 μM down to <50 nM in multiple cell lines from multiple species. Furthermore, we show that our compounds increase intracellular granulins in cell lines as well as in iPSC-derived neurons from FTD-*GRN* patients. Conversely, blocking the cell surface PGRN receptor sortilin with a monoclonal antibody reduces granulins in neurons. Functionally, these compounds reduce lipid droplet accumulation brought about by PGRN deficiency and reduce LPS-induced pro-inflammatory cytokine release in a concentration-dependent fashion. In conclusion, our approach yielded novel small molecules that have favorable drug-like properties and are well-suited for further development for the treatment of FTD-*GRN*.

Results:

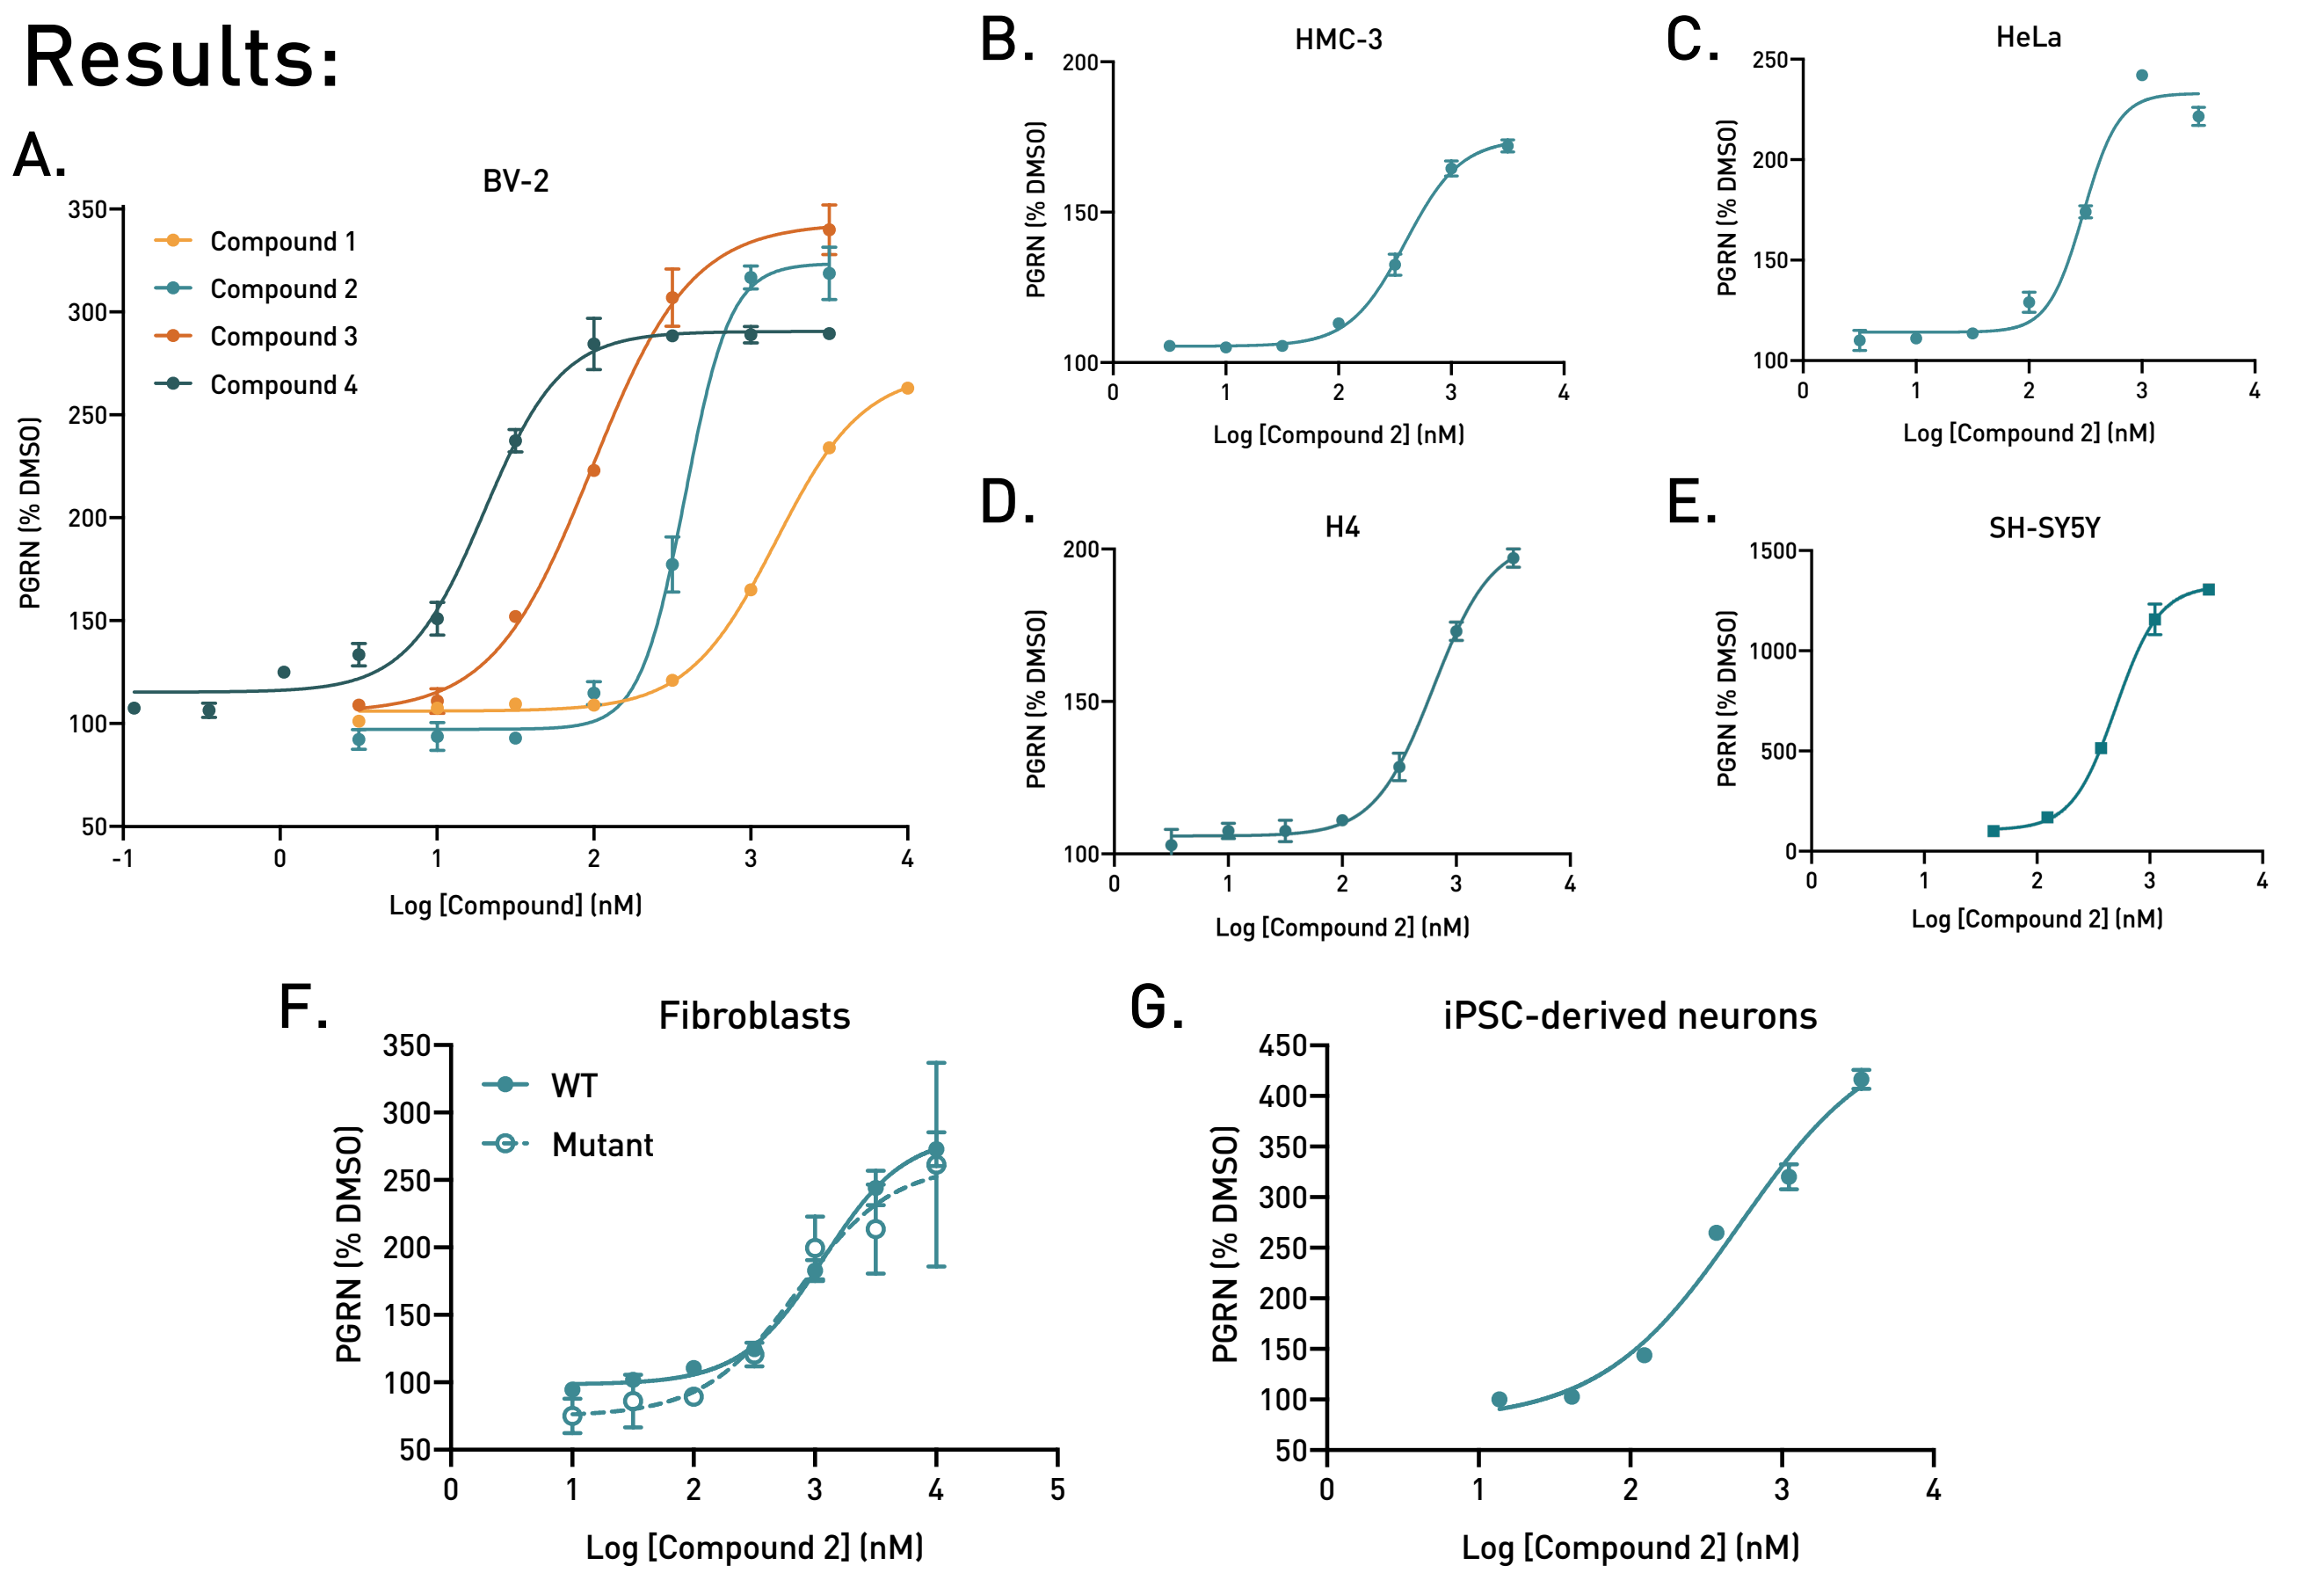
A.
BV-2
Compound 1
Compound 2
Compound 3
Compound 4
PGRN (% DMSO)
Log [Compound] (nM)
B.
HMC-3
C.
HeLa
D.
H4
E.
SH-SY5Y
Log [Compound 2] (nM)
F.
Fibroblasts
WT
Mutant
G.
iPSC-derived neurons

Figure 1. Novel small molecules increase PGRN secretion in a variety of cell lines/types. (A) Primary screen is performed in BV-2 cells (mouse microglia). Potency improvement is shown for 4 compounds out of >800 screened. **(B-E)** Compound 2 potency tracks across species and multiple human cell lines of different lineage **(B)** Embryonic microglia. **(C)** Cervical cancer. **(D)** Neuroglioma. **(E)** Neuroblastoma. **(F-G)** Compound 2 also increases PGRN secretion from **(F)** WT and *GRN* mutant human fibroblasts as well as **(G)** *GRN* mutant human iPSC-derived neurons.

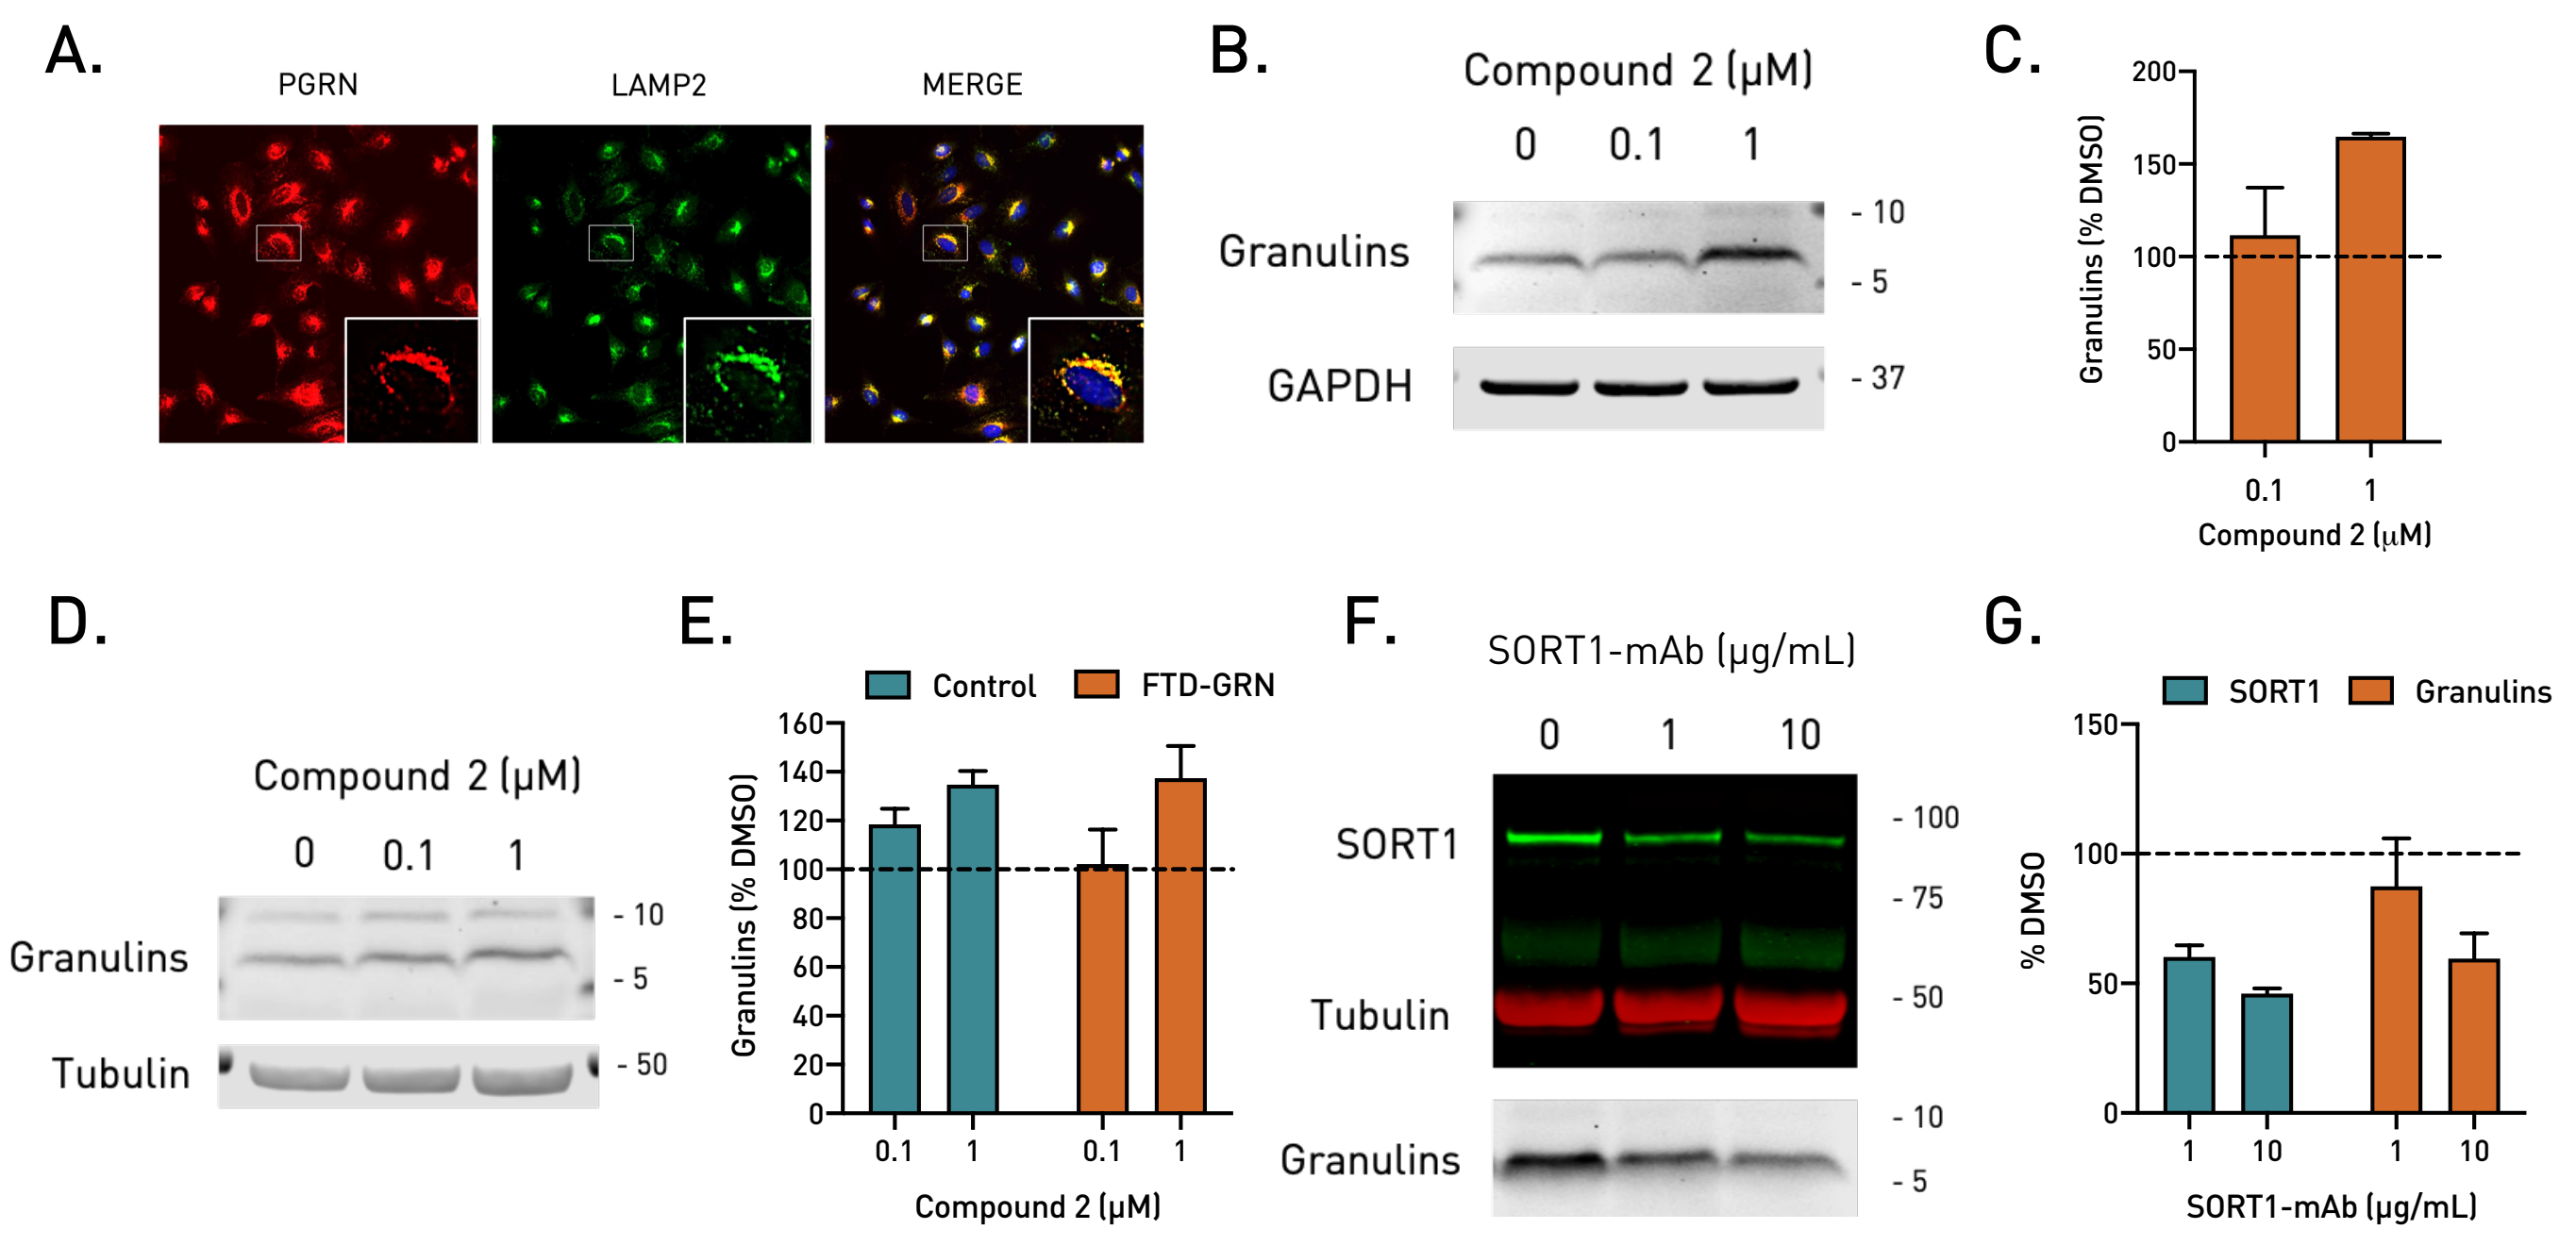
A.
PGRN
LAMP2
MERGE
B.
Compound 2 (μM)
0 0.1 1
Granulins
GAPDH
- 10
- 5
- 37
C.
Granulins (% DMSO)
Compound 2 (μM)
D.
Compound 2 (μM)
0 0.1 1
Granulins
Tubulin
- 10
- 5
- 50
E.
Control
FTD-GRN
Granulins (% DMSO)
Compound 2 (μM)
F.
SORT1-mAb (μg/mL)
0 1 10
SORT1
Tubulin
Granulins
- 100
- 75
- 50
- 10
- 5
G.
SORT1
Granulins
% DMSO
SORT1-mAb (μg/mL)

Figure 2. Compound 2 increases lysosomal granulins in cultured cell lines and iPSC-derived neurons. (A) PGRN/granulins co-localize with LAMP2-positive lysosomes in HeLa cells; 20X magnification. **(B)** Compound 2 (24 hr treatment) increases lysosomal granulins in SH-SY5Y cells; quantified in **(C)**. **(D)** Compound 2 increases lysosomal granulins in control (shown) and FTD-*GRN* iPSC-derived neurons; quantified in **(E)**. **(F)** Sortilin-blocking monoclonal antibody (SORT1-mAb) decreases sortilin and lysosomal granulins in iPSC-derived neurons in a concentration-dependent manner; quantified in **(G)**.

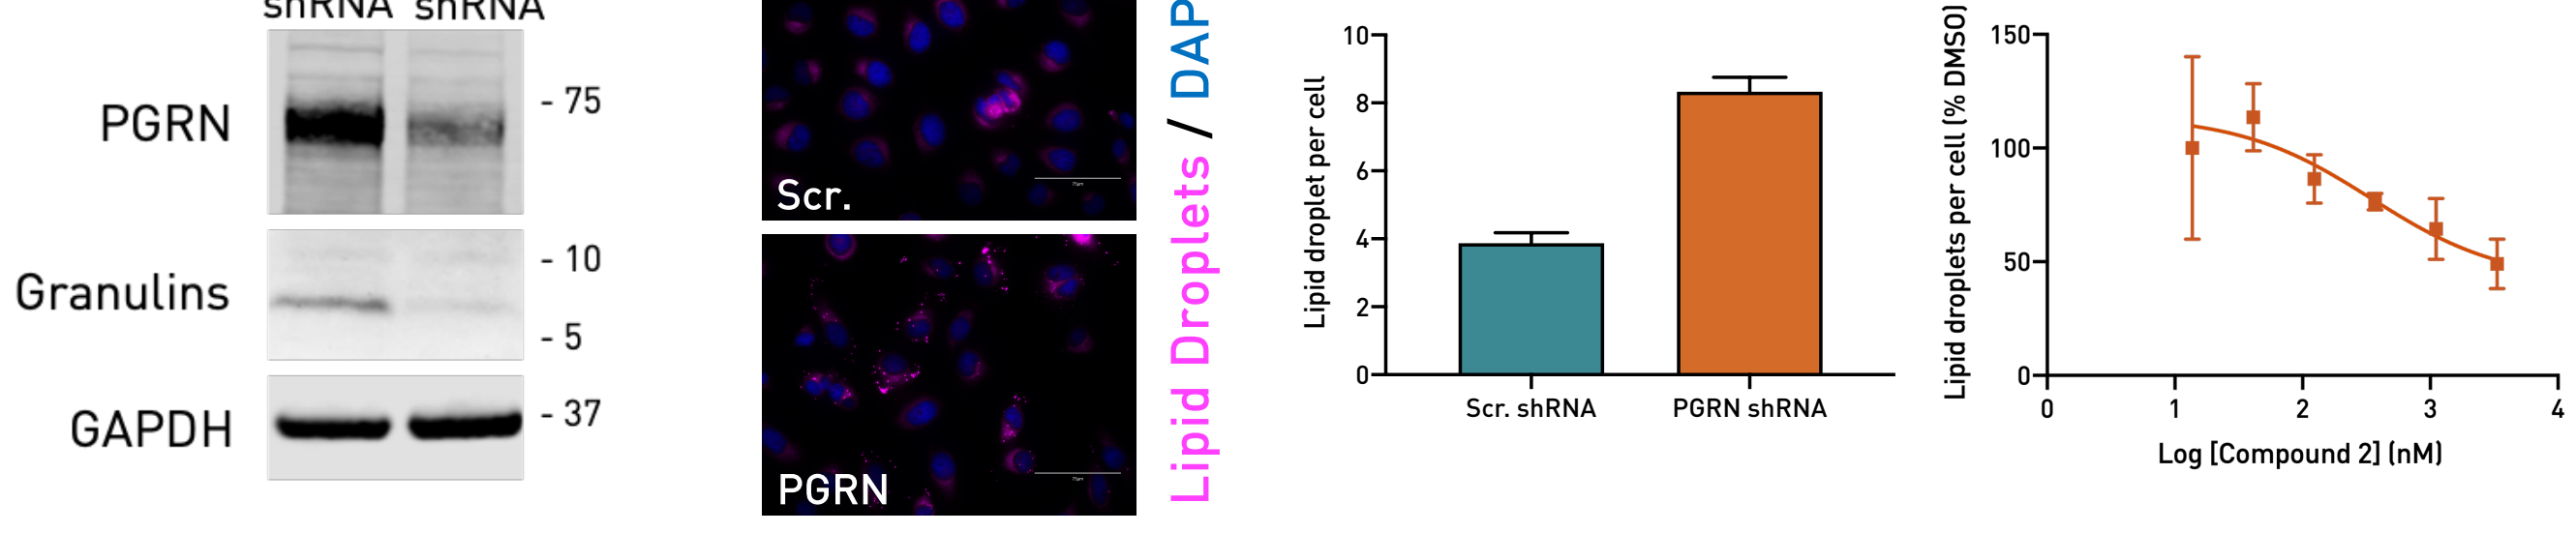
A.
Scr shRNA
PGRN shRNA
PGRN
Granulins
GAPDH
- 75
- 10
- 5
- 37
B.
Scr.
PGRN
Lipid Droplets / DAPI
C.
Lipid droplet per cell
Scr. shRNA
PGRN shRNA
D.
Lipid droplets per cell (% DMSO)
Log [Compound 2] (nM)

Figure 3. Compound 2 reduces lipid droplet accumulation in PGRN deficient cells. (A) PGRN shRNA leads to decreased PGRN and granulins. **(B)** Lipid droplets (LD; LipidSpot 610) are increased in PGRN deficient HeLa cells; 40X. **(C)** High-content image analysis reveals a 2-fold increase in LDs which can be reversed in a concentration-dependent manner by compound 2 **(D)**.

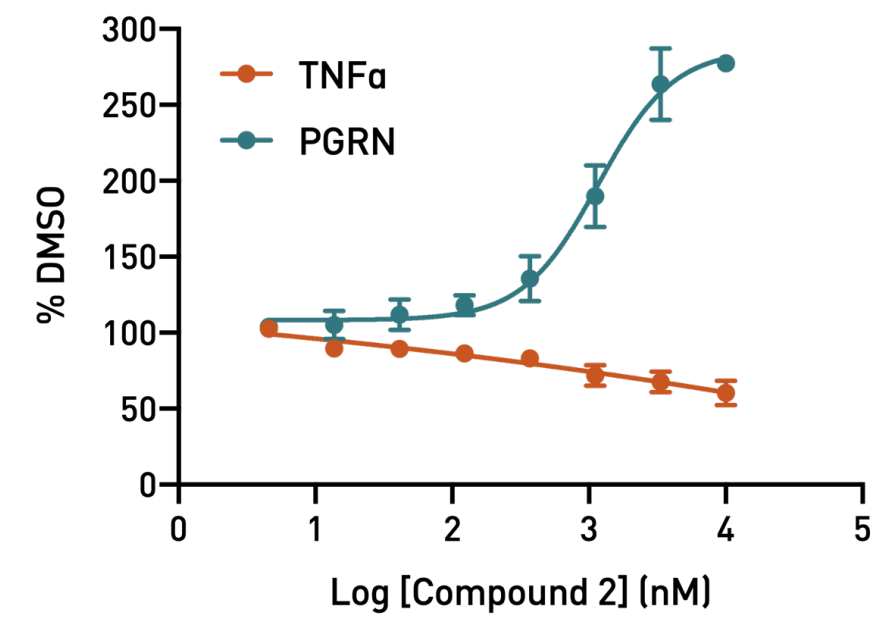
TNFα
PGRN
% DMSO
Log [Compound 2] (nM)

Figure 4. Compound 2 reduces LPS-induced TNFα secretion. BV-2 cells are co-treated with 5 μg/ml LPS and different concentrations of Compound 2 for 16 hours. Secreted PGRN and TNFα are measured by ELISA.

Conclusions:

- A series of small molecules were screened, and SAR optimization led to a highly potent, novel set of PGRN-enhancing compounds.
- Compounds increase PGRN secretion and lysosomal granulins in multiple relevant cell lines.
- Sortilin mAb decreases lysosomal granulins.
- Compounds reverse lipid droplet accumulation due to PGRN deficiency.
- Compounds modulate a neuroinflammatory marker.

Disease

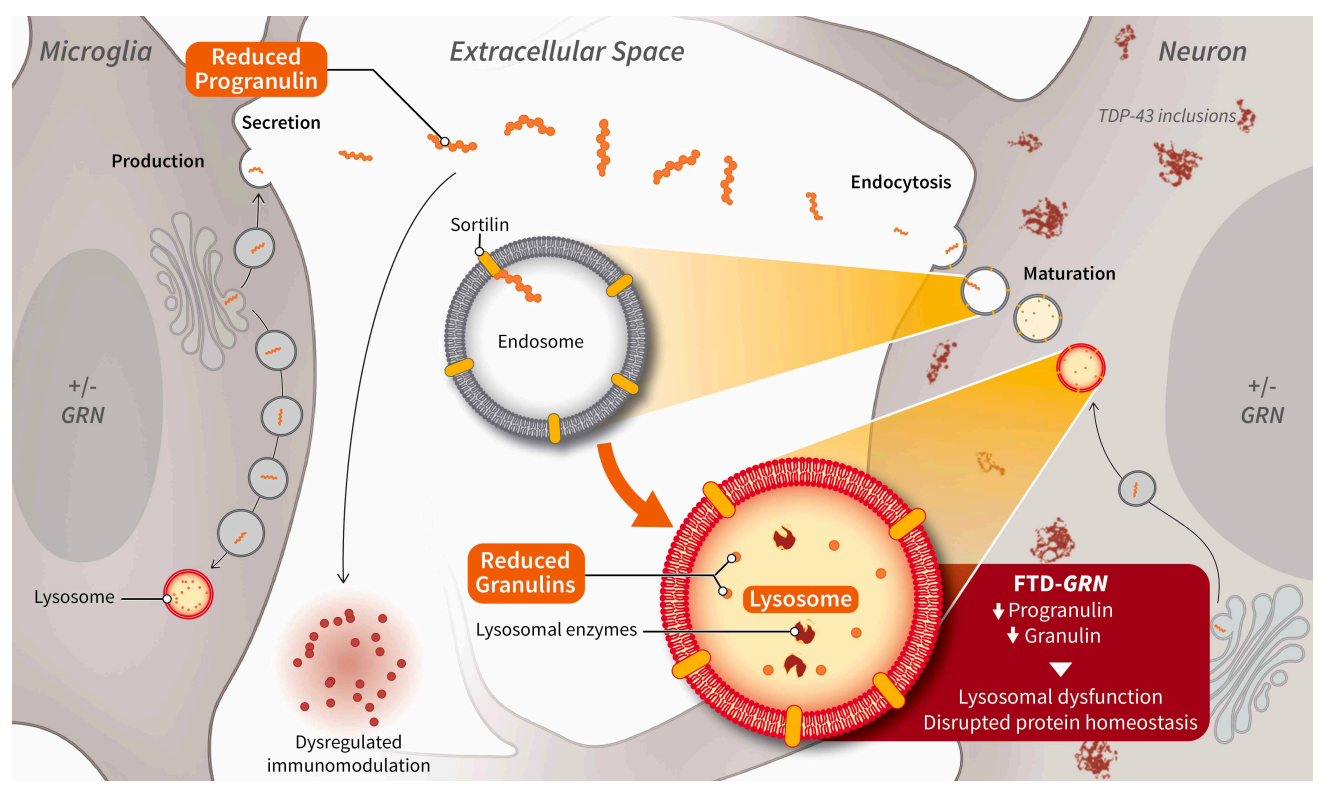
Microglia
Reduced Progranulin
Extracellular Space
Neuron
Secretion
Production
TDP-43 inclusions
Endocytosis
Sortilin
Maturation
Endosome
+/- GRN
+/- GRN
Reduced Granulins
Lysosomal enzymes
Lysosome
Lysosome
FTD-GRN
Progranulin
Granulin
Lysosomal dysfunction
Disrupted protein homeostasis
Dysregulated immunomodulation

Treatment Goal

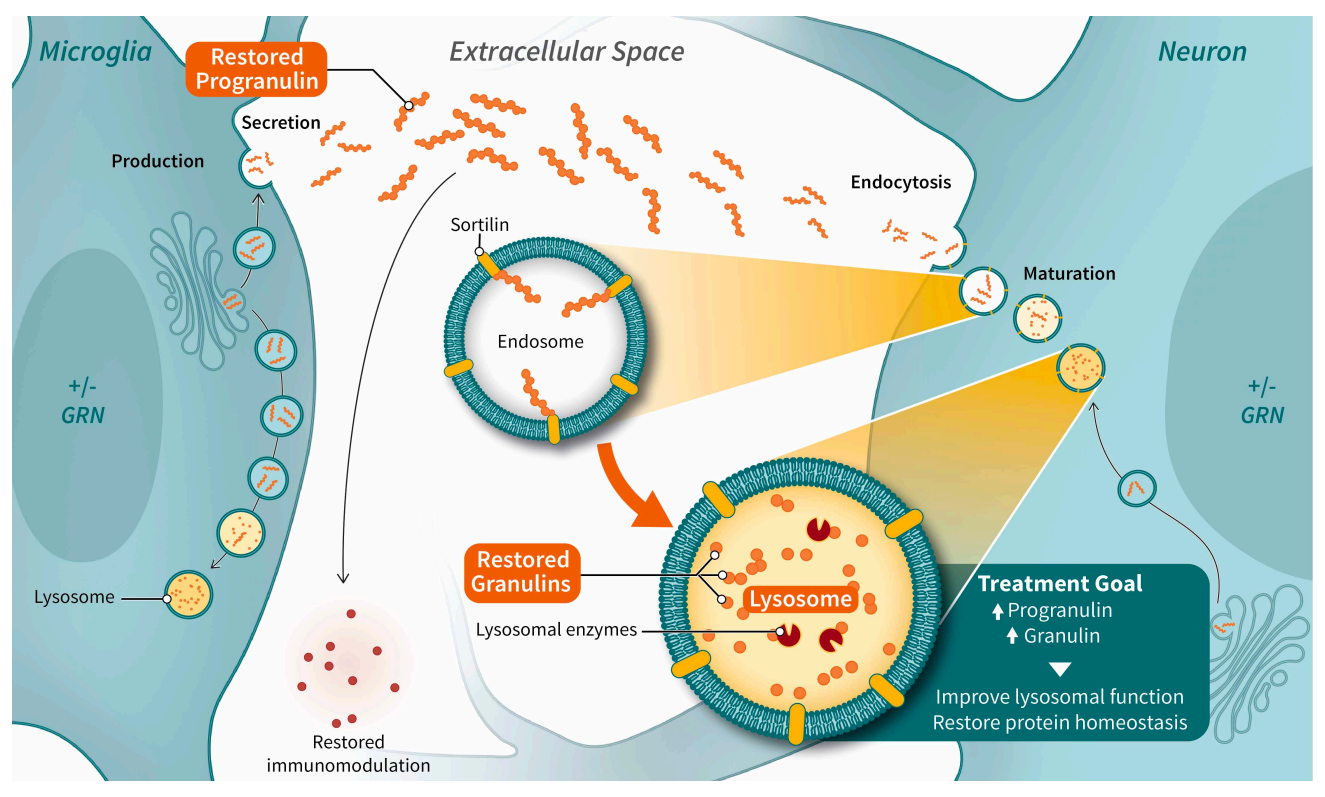
Microglia
Restored Progranulin
Extracellular Space
Neuron
Secretion
Production
Endocytosis
Sortilin
Maturation
Endosome
+/- GRN
+/- GRN
Restored Granulins
Lysosomal enzymes
Lysosome
Lysosome
Treatment Goal
Progranulin
Granulin
Improve lysosomal function
Restore protein homeostasis
Restored immunomodulation

Contacts: choller@arkudatx.com, jfblain@arkudatx.com @ArkudaTx