ARKD-104, A Potential Treatment of Frontotemporal Dementia and Other Neurodegenerative Disorders

Hung, S., Hurst, R. S., Blain, J-F., Holler, C., Chen, A., Brand, M., Alves, A-M., Astarita, G., Gallegos, R., Greenberg, G., Henry, S., Koenig, G., McTighe, S.M., Peng, X., Ross, J., Thompson, B. M., Watters, K., Wozniak, M., Lanter, J. C., Burnett, D. A.

Arkuda Therapeutics, Watertown, Massachusetts, United States

Background:
Heterozygous mutations in the gene encoding progranulin (*GRN*) that lead to haploinsufficiency of the progranulin protein (PGRN) cause the fatal neurodegenerative disease frontotemporal dementia (FTD-*GRN*). A promising treatment approach for FTD-*GRN* is to restore PGRN to levels detected in healthy individuals. In pursuit of this objective, Arkuda Therapeutics is developing brain-penetrant, orally bioavailable small molecules that increase PGRN levels within the central nervous system (CNS).

Method:
A functional cell-based assay was utilized to identify novel compounds that stimulate PGRN secretion from a murine microglial cell line. Compounds with good *in vitro* potency were further optimized through an iterative series of selectivity, ADME, pharmacokinetic (PK), safety and pharmacology assessments.

Result:
When applied to cells, ARKD-104 increased PGRN and PSAP secretion and engaged lysosomal pathways as evidenced by intracellular changes in granulins, saposins, and lysosomal lipids including bis(monoacylglycero)phosphate (BMP). Oral administration of ARKD-104 to cynomolgus monkeys resulted in a dose and exposure-dependent increase in PGRN measured in cerebrospinal fluid (CSF). The exposure-response relationship demonstrated that a systemic exposure of 8 nM ARKD-104 in plasma (unbound) doubled the amount PGRN in CSF. PGRN levels were further increased to more than 4-times pre-dose levels at the highest tested dose. Elevation of PGRN levels was sustained with repeat dosing of ARKD-104. ARKD-104 was characterized for CNS drug-like properties including PK, metabolism, brain penetration, selectivity, and safety. In these assessments, ARKD-104 demonstrated a profile suggesting that beneficial increases in PGRN can be achieved with once daily oral dosing in human.

Conclusion:
ARKD-104 has excellent CNS drug-like properties, increases PGRN in the CNS of non-human primates and increases important cofactors of lysosomal enzymes, . ARKD-104 is therefore a promising candidate for continued development as a treatment for FTD-*GRN*, Parkinson's disease and other neurodegenerative disorders exhibiting lysosomal dysfunction.

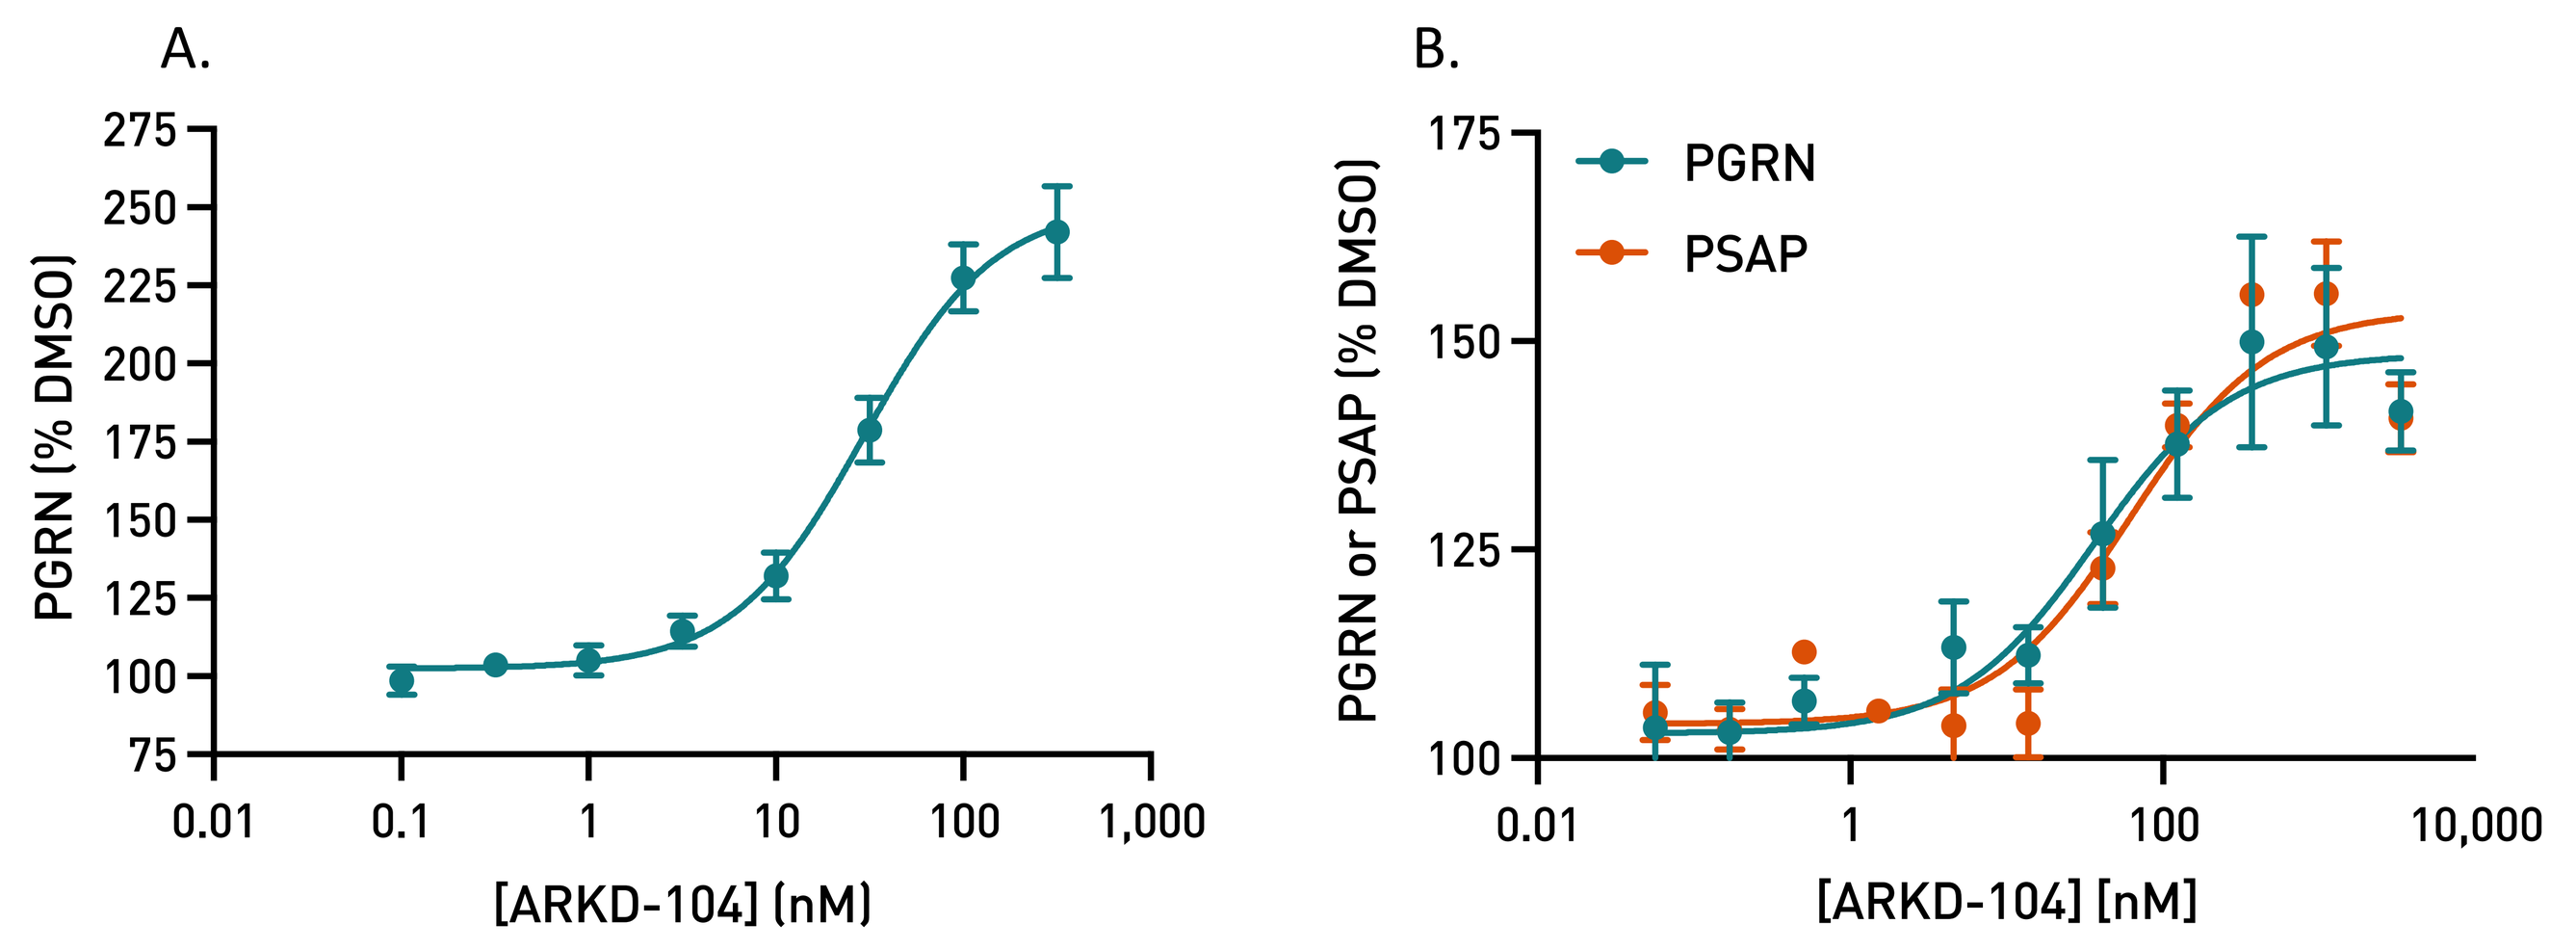
A.
PGRN (% DMSO)
[ARKD-104] (nM)
B.
PGRN
PSAP
PGRN or PSAP (% DMSO)
[ARKD-104] (nM)

Figure 1. ARKD-104 increases PGRN and PSAP secretion.
A. BV-2 mouse microglial cells and **B.** RAW264.7 mouse macrophages were treated with ARKD-104 for 16 hours. Secreted PGRN and PSAP were measured in the media by ELISA.

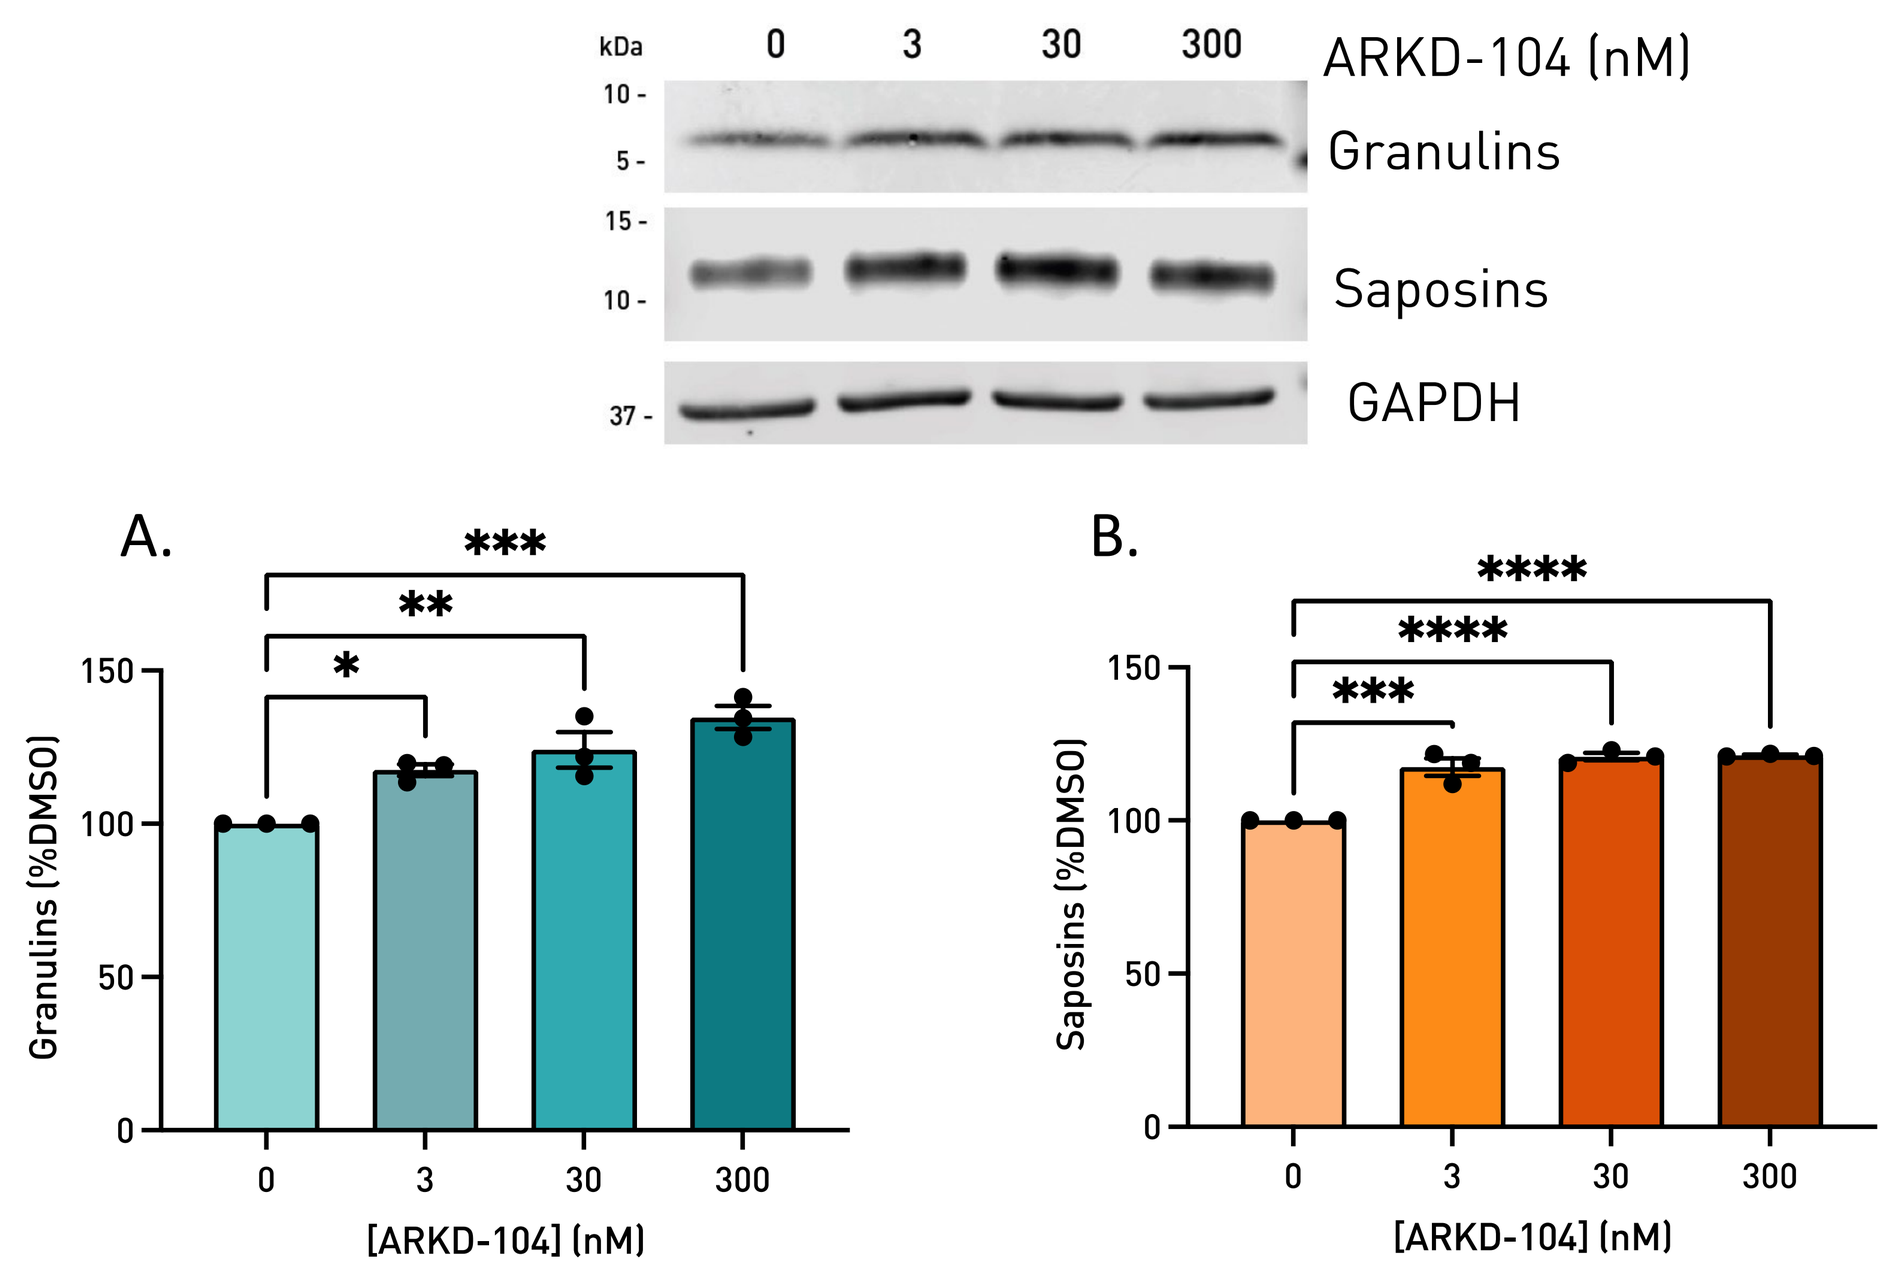
ARKD-104 (nM)
Granulins
Saposins
GAPDH
A.
Granulins (%DMSO)
B.
Saposins (%DMSO)
[ARKD-104] (nM)

Figure 2. ARKD-104 increases lysosomal granulins and saposins.
SH-SY5Y human neuroblastoma cells were treated for 4 hours. **A.** Granulins and **B.** saposins were measured by Western blot.

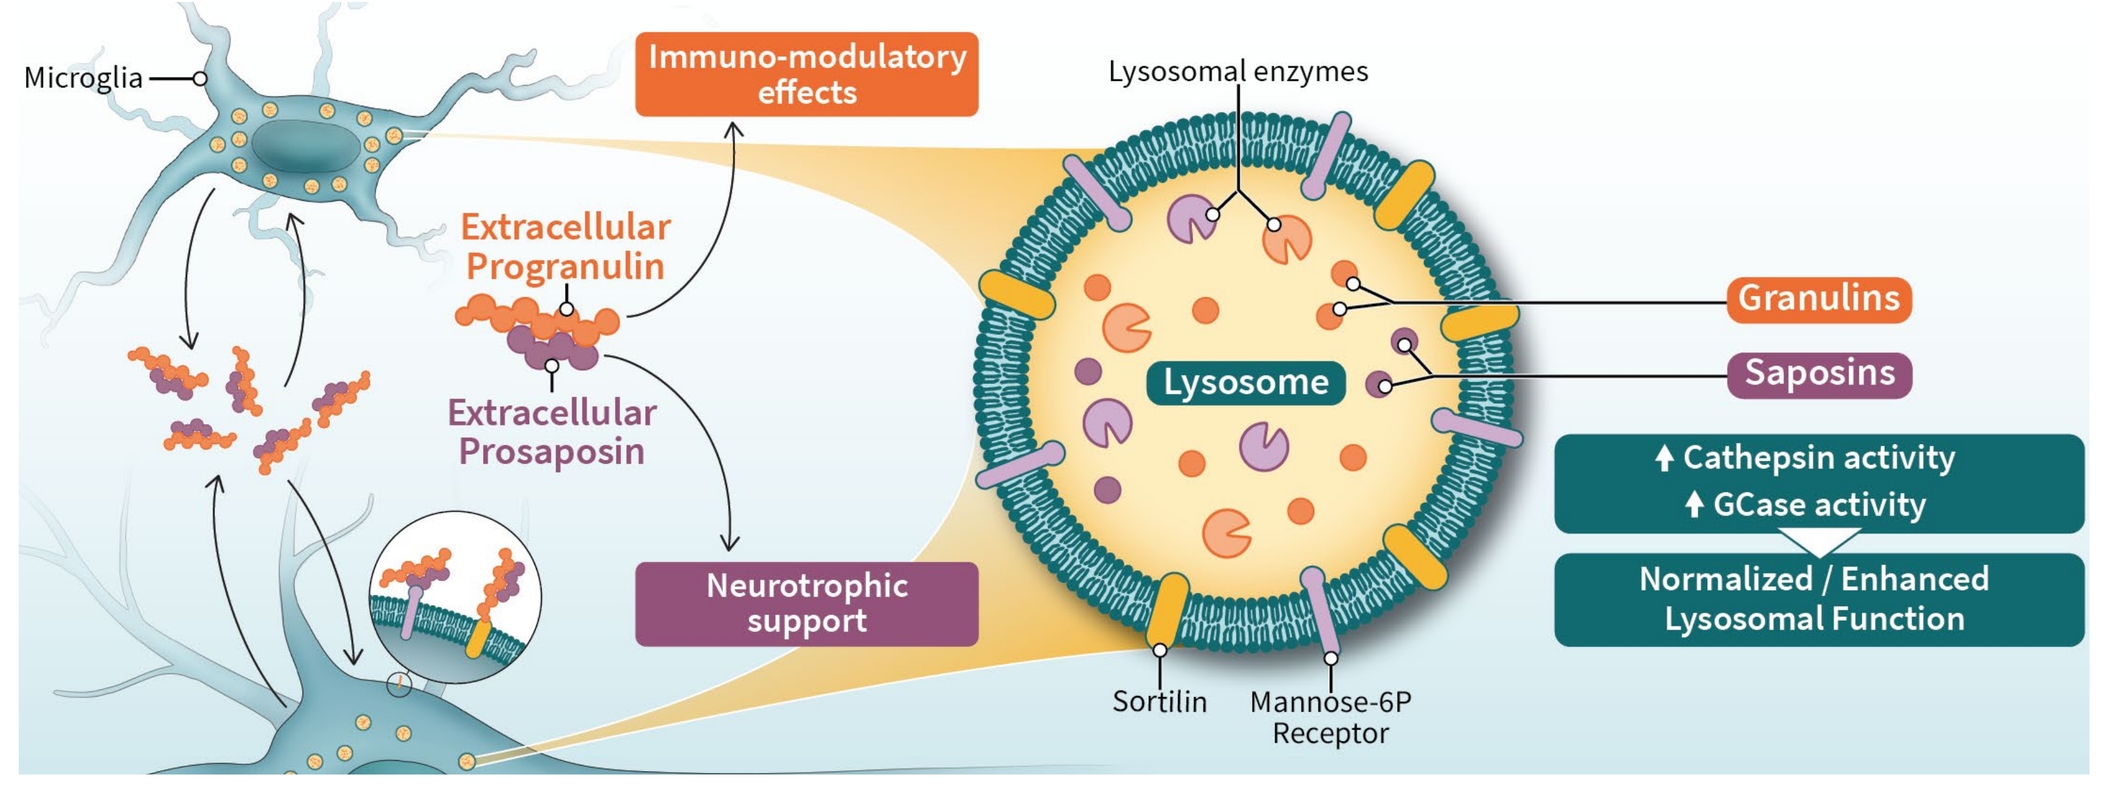
Microglia
Immuno-modulatory effects
Extracellular Progranulin
Extracellular Prosaposin
Neurotrophic support
Lysosomal enzymes
Lysosome
Granulins
Saposins
Cathepsin activity
GCase activity
Normalized / Enhanced Lysosomal Function
Sortilin
Mannose-6P Receptor

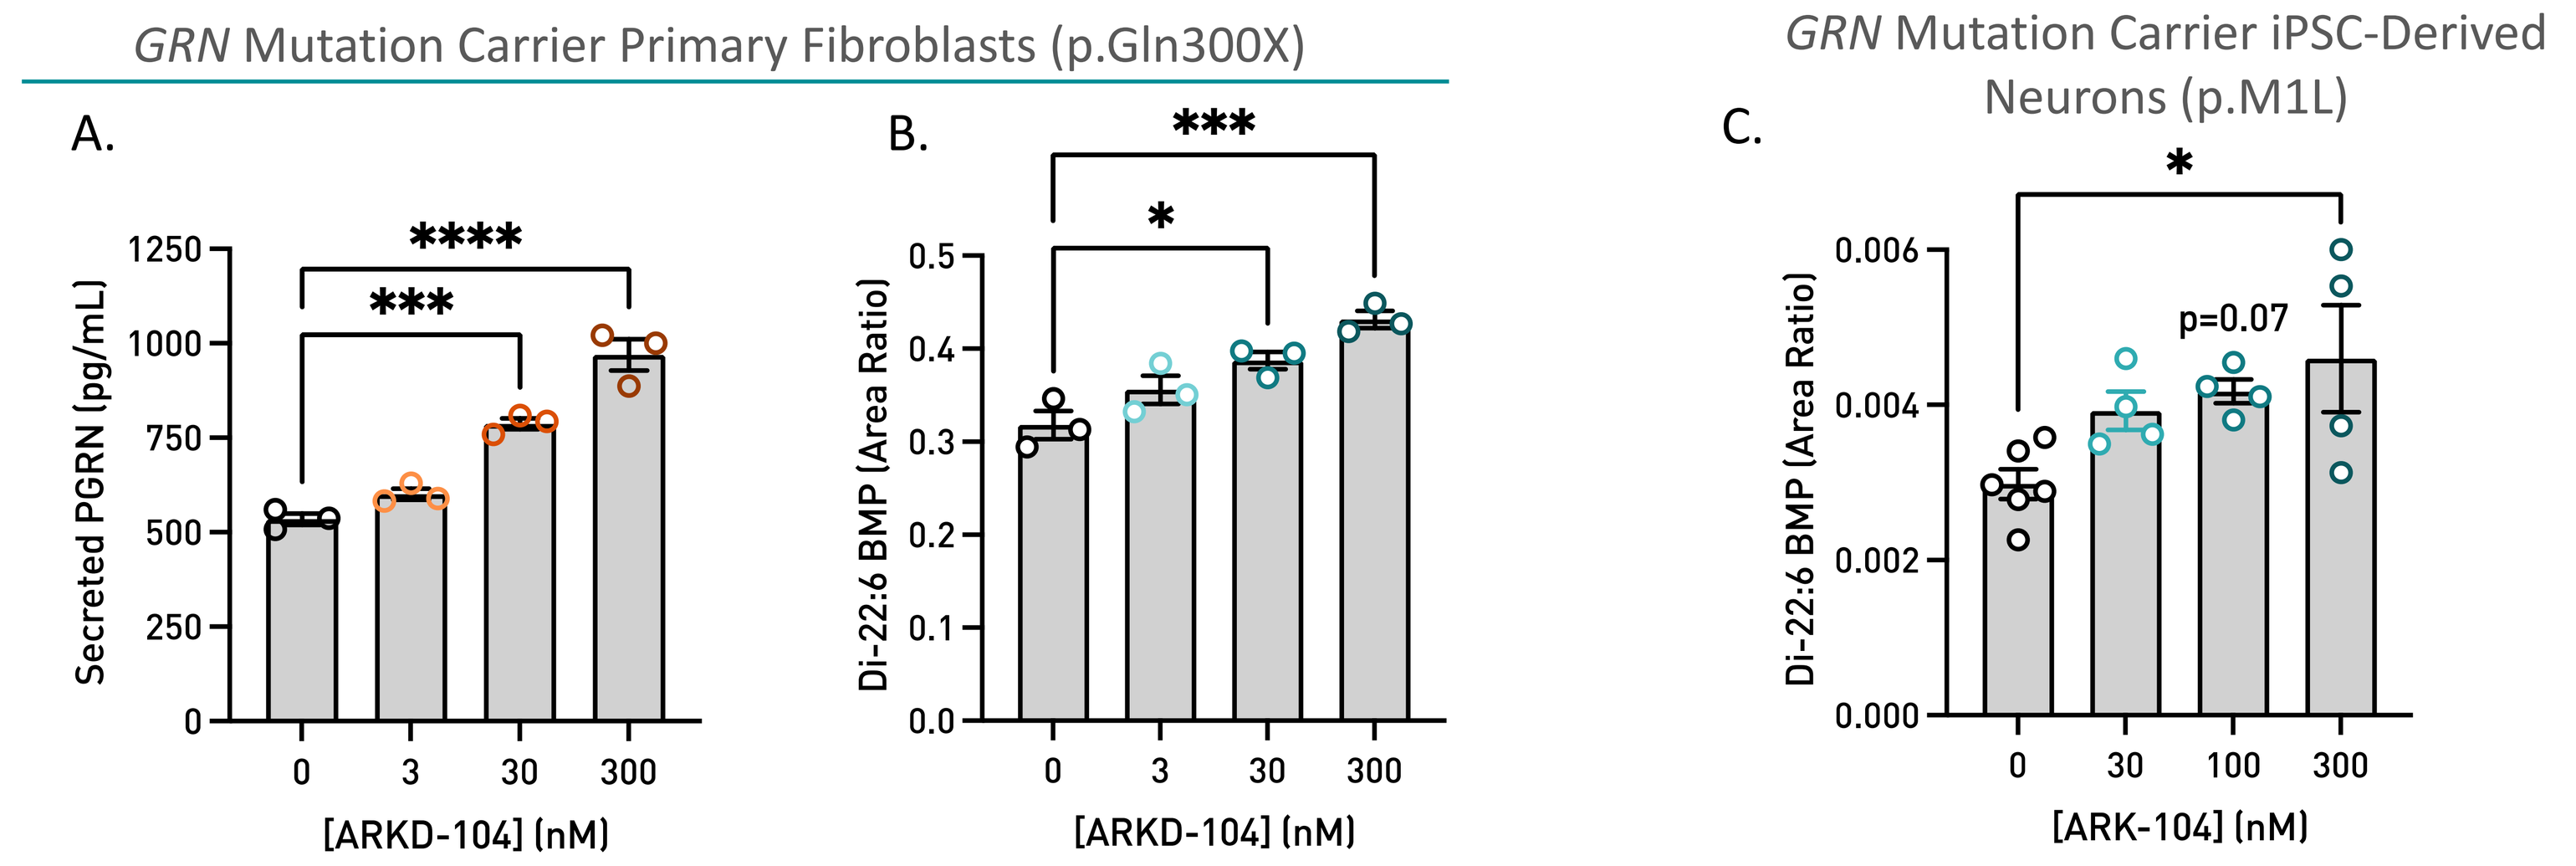
GRN Mutation Carrier Primary Fibroblasts (p.Gln300X)
A.
Secreted PGRN (pg/mL)
[ARKD-104] (nM)
B.
Di-22:6 BMP (Area Ratio)
[ARKD-104] (nM)
GRN Mutation Carrier iPSC-Derived Neurons (p.M1L)
C.
p=0.07
Di-22:6 BMP (Area Ratio)
[ARK-104] (nM)

Figure 3. ARKD-104 engages the lysosome as demonstrated by the increase in the lysosomal lipid BMP. Human dermal fibroblasts from a *GRN* mutation carrier were treated with ARKD-104 at different concentrations for 24 hours. **A.** Secreted PGRN was measured by ELISA and **B.** di-22:6 BMP was quantified by LC/MS. The same treatment was applied to FTD-*GRN* iPSC-derived neurons and di-22:6 BMP (**C.**) was quantified by LC/MC. The changes observed suggest lysosome engagement. One-way ANOVA with multiple comparisons * p<0.05; *** p< 0.001; **** p<0.0001.

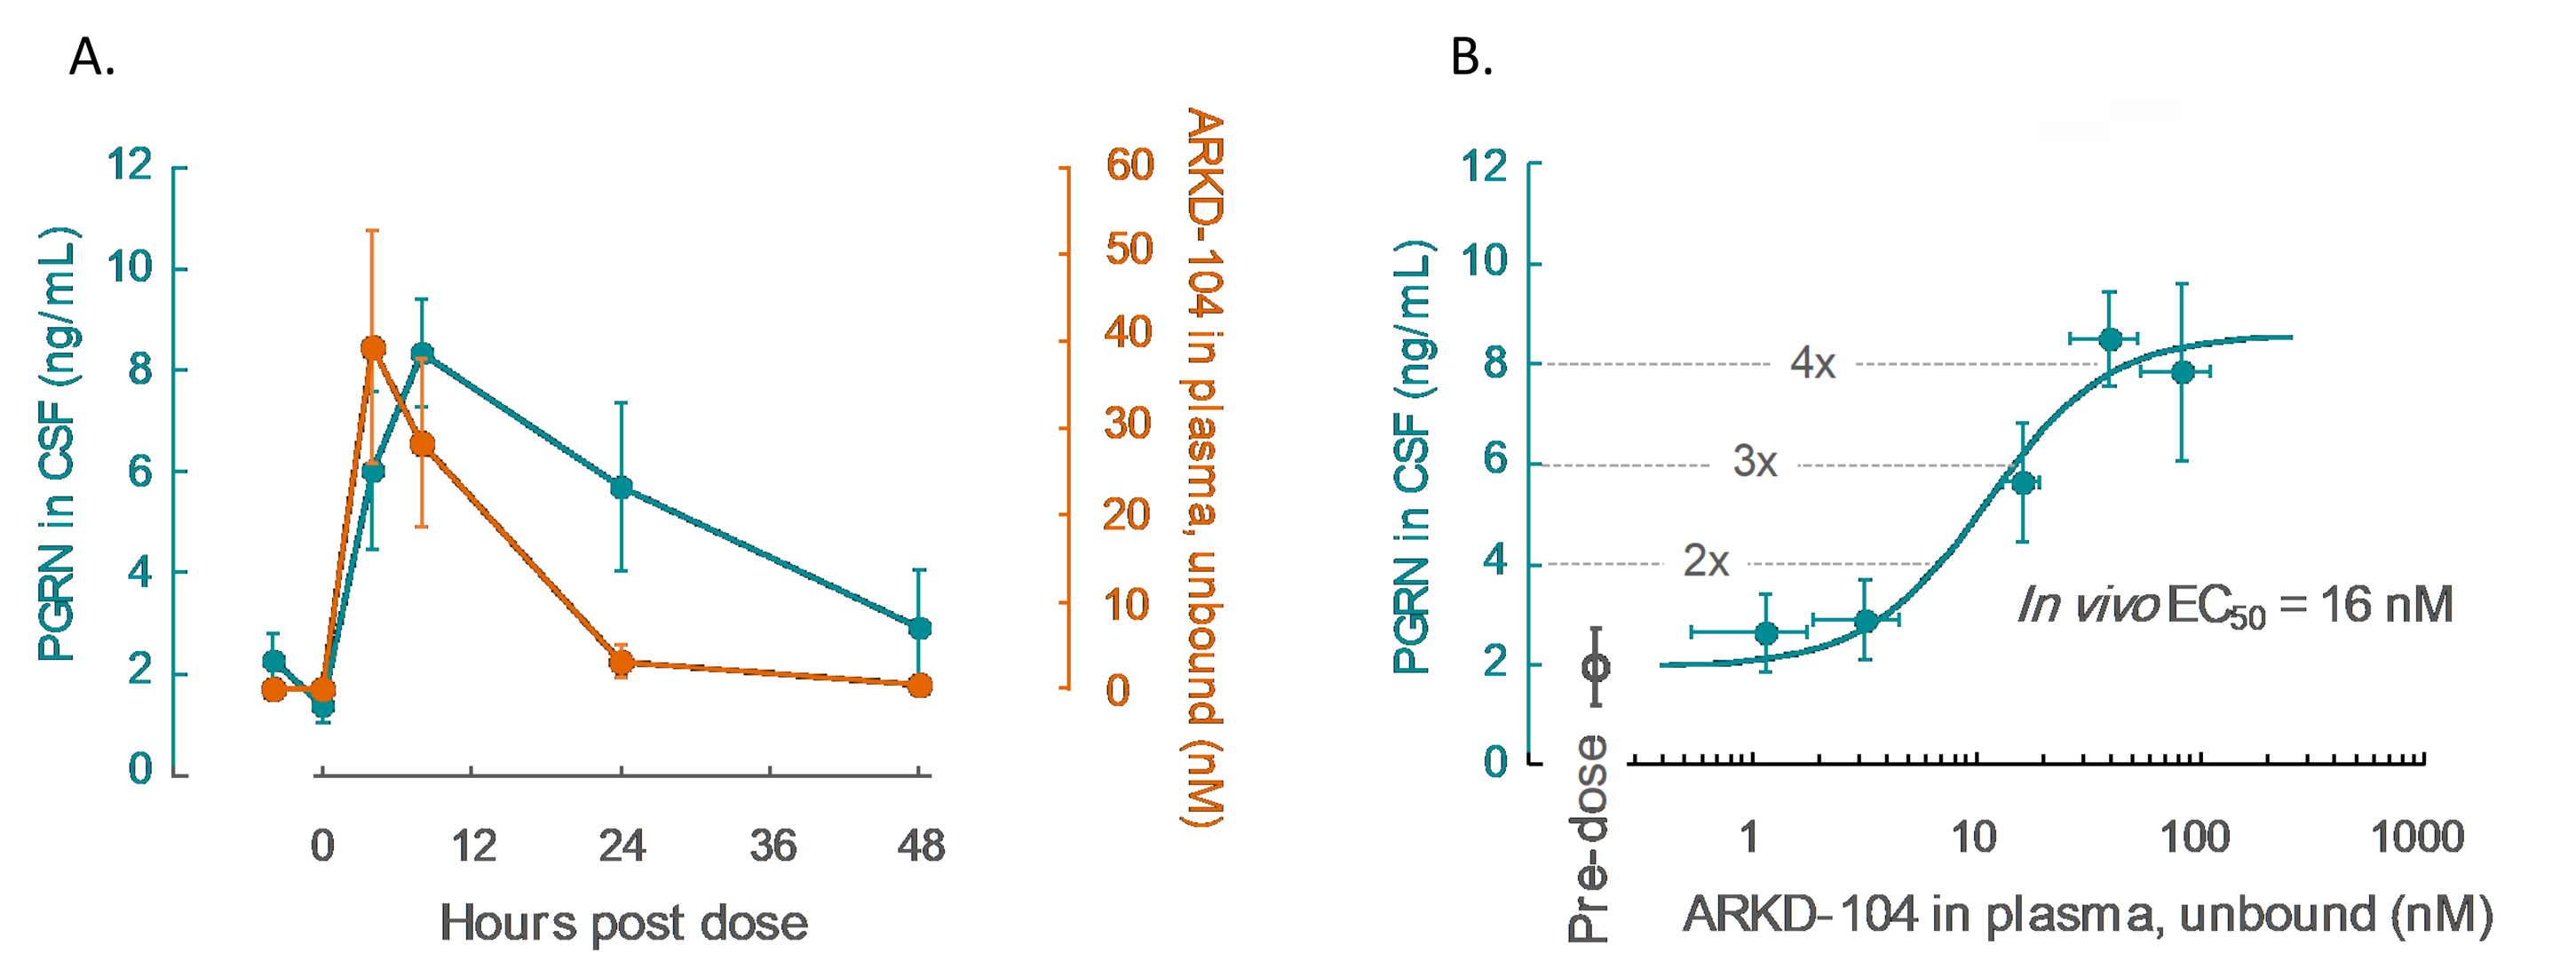
A.
PGRN in CSF (ng/mL)
ARKD-104 in plasma, unbound (nM)
Hours post dose
B.
PGRN in CSF (ng/mL)
4x
3x
2x
In vivo EC50 = 16 nM
Pre-dose
ARKD-104 in plasma, unbound (nM)

Figure 4. ARKD-104 leads to a dose-dependent increase in CSF PGRN. CSF was sampled from cisterna magna-ported cynomologus monkeys (n = 6) before dosing to establish PGRN levels baseline (Pre-Dose average: 1.92 ± 0.86 ng/mL). ARK-1252 was administered orally at 0.5, 2, 5, 10, and 20 mg/kg and CSF was sampled 4-, 8-, 24-, and 48-hours post-dose. **A.** Time course of PK/PD for PGRN changes at 10 mg/kg. **B.** Exposure-response plot was generated by plotting the maximal PGRN observed per animal, per dose against the Cmax of ARKD-104 measured in CSF by LC/MS. The *in vivo* exposure-response data were fit by a logistic function to derive an *in vivo* EC_{50} of 16 nM

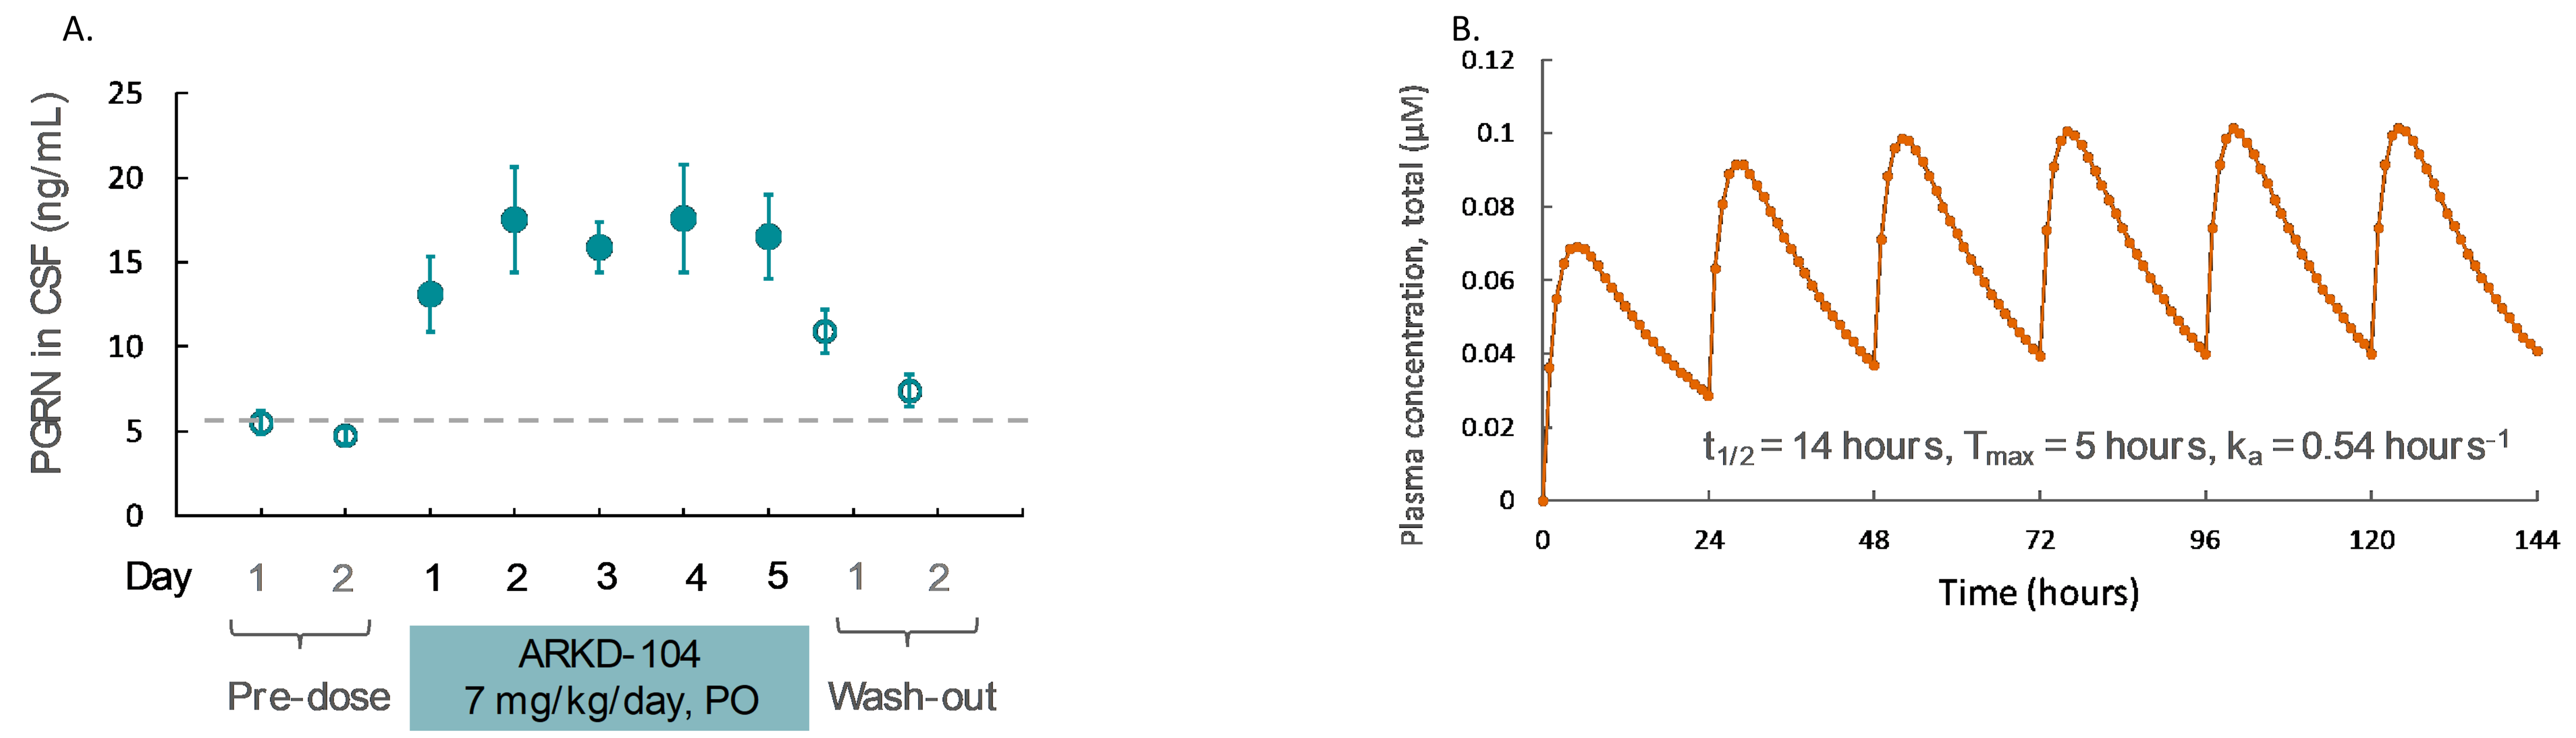
A.
PGRN in CSF (ng/mL)
Day
Pre-dose
ARKD-104
7 mg/kg/day, PO
Wash-out
B.
Plasma concentration, total (µM)
t1/2 = 14 hours, Tmax = 5 hours, ka = 0.54 hours-1
Time (hours)

Figure 5. PGRN-inducing properties of ARKD-104 do not change following repeat dosing in NHP. Cisterna magna-ported cynomologus monkeys (n = 6) were used for this study. CSF was sampled 24-, and 0-hours before dosing to establish baseline PGRN levels. Compound was administered orally at 7 mg/kg daily for 5 days and CSF was sampled 8-hours post-dose as well as 24- and 48-hours post-last dose. CSF compound concentration was measured by LC/MS. **A.** PGRN levels increased and stayed above 2x baseline after each dose. PGRN returned to baseline levels as compound is cleared from the CSF. **B.** Simulated human pharmacokinetic profile following repeated oral daily administration of ARKD-104.

Conclusions

- ARKD-104 increases the secretion of PGRN with an unbound EC_{50} of 18 nM as well as PSAP and increases both lysosomal granulins and saposins at similar concentrations *in vitro.*
- ARKD-104 modulates lipids (BMP and TG) in FTD-*GRN* neurons and fibroblasts suggesting biological engagement of the lysosome.
- ARKD-104 produces a dose-dependent increase in CSF PGRN in non-human primates and has an *in vivo* EC_{50} of 16 nM.
- ARKD-104 can be dosed for multiple days without loss in pharmacodynamic effect.
- Based on its overall profile, ARKD-104 shows great potential for the treatment of FTD-*GRN,* as well as other neurodegenerative diseases currently under investigation.
- ARKD-104 undergoing IND-enabling studies; anticipate first-in-human trial 1H 2024.